#673

Impact of Number of Lines of Therapy Following First-Line Immuno-oncology Combination on Overall Survival in Patients with Metastatic Renal Cell Carcinoma

Audreylie Lemelin[1], Matthew S. Ernst[1], J. Connor Wells[2], Vishal Navani[1], Bradley Alexander McGregor[3], Shirley S. Wong[4], Sumanta K. Pal[5], Naveen S. Basappa[6], Anil Kapoor[7], Jae-Lyun Lee[8], Frede Donskov[9,10], Haoran Li[11], Takeshi Yuasa[12], Rose Chang[13], Lynn Huynh[13], Catherine Nguyen[13], Ashley Holub[13], Louise Clear[13], Mei Sheng Duh[13], and Daniel Yick Chin Heng[1]

[1]Tom Baker Cancer Centre, University of Calgary, Calgary, Canada; [2]BC Cancer Agency, Vancouver, Canada; [3]Dana-Farber Cancer Institute, Boston, United States; [4]Western Health, University of Melbourne, Melbourne, Australia; [5]City of Hope Comprehensive Cancer Center, Duarte, United States; [6]Cross Cancer Institute, University of Alberta, Edmonton, Canada; [7]Juravinski Cancer Centre, McMaster University, Hamilton, Canada; [8]Asan Medical Center, University of Ulsan College of Medicine, Seoul, Republic of Korea; [9]Aarhus University Hospital, Aarhus, Denmark; [10]Southern Denmark University Hospital, Esbjerg, Denmark; [11]Huntsman Cancer Institute, University of Utah, Salt Lake City, United States; [12]Cancer Institute Hospital, Japanese Foundation for Cancer Research, Tokyo, Japan; [13]Analysis Group Inc, Boston, United States

BACKGROUND

- While immuno-oncology-based combination therapies are the current standard of care for patients with metastatic renal cell carcinoma (mRCC), patients around the world may receive a range of different treatment sequences and lines of therapy (LOTs)[1]
- The regimen received in first-line (1L) impacts the choice for subsequent LOTs. A sequence strategy example: patients receiving 1L nivolumab plus ipilimumab (Nivo+Ipi) may be treated with second-line (2L) and third-line (3L) vascular endothelial growth factor receptor targeted kinase inhibitors (VEGFR-TKI), whereas patients receiving 1L pembrolizumab or avelumab with axitinib (IO+Axi) may be treated with only one subsequent 2L VEGFR-TKI[2,3]
- There is limited information on the optimal treatment sequences for patients with mRCC treated with 1L immuno-oncology (IO) combination therapy and its impact on clinical decisions regarding treatment sequencing and overall clinical outcomes

METHODS

Study population

- Adult mRCC patients who received ≥3 LOTs starting with 1L Nivo+Ipi or ≥2 LOTs starting with 1L IO+Axi from International Metastatic RCC Database Consortium (IMDC) centers in 2014-2022 were included in this real-world, retrospective database study

Study outcomes

- Overall survival (OS): time from 1L IO combination treatment initiation to death
- Time to treatment discontinuation (TTD): time from treatment initiation to discontinuation due to any reason
- Time to next treatment (TTNT): time from treatment initiation to subsequent treatment initiation

Statistical analysis

- Patient characteristics prior to 1L treatment initiation and 1L-3L treatment patterns were described for the ≥3 LOTs starting with 1L Nivo+Ipi and ≥2 LOTs starting with 1L IO+Axi cohorts using the mean (standard deviation [SD]) and median values for continuous variables and frequency distributions for categorical variables
 - Differences between cohorts were compared using Pearson chi-squared tests (or Fisher's exact test as appropriate) for categorical variables, while continuous variables were compared using Wilcoxon rank-sum tests
- For all study outcomes, Kaplan-Meier was used to estimate median time to event and 95% confidence intervals (CIs). OS was stratified by IMDC risk group
 - To account for potential guaranteed time bias, landmark analyses for OS were conducted at 12, 18, and 24 months following 1L initiation among patients who were still alive and being followed. As a sensitivity analysis, OS was computed for patients in the ≥2 LOTs starting with 1L IO+Axi cohort who received any 3L
- Hazard ratios (HR) and 95% CIs were estimated using Cox proportional hazards models to compare OS between cohorts while adjusting for baseline covariates

RESULTS

- Patient characteristics [**Table 1**]
 - Overall, 128 patients were treated ≥3 LOTs starting with 1L Nivo+Ipi while 104 patients were treated with ≥2 LOTs starting with 1L IO+Axi
 - Patients included had a median age of 61.5 years and were predominantly White and male
- Treatment patterns [**Figure 1**]
 - For the ≥3 LOTs starting with 1L Nivo+Ipi cohort, the most common 2L treatments were sunitinib (38%), cabozantinib (27%), and pazopanib (20%)
 - For the ≥2 LOTs starting with 1L IO+Axi cohort, the most common 2L treatments were cabozantinib (57%) and sunitinib (10%)
- TTD and TTNT [**Table 2**]
 - TTD and TTNT were numerically longer across LOTs for patients in the ≥2 LOTs starting with 1L IO+Axi cohort compared with the ≥3 LOTs starting with 1L Nivo+Ipi cohort
- OS
 - Median OS was 33.0 months [95% CI: 27.6, 40.2] for the ≥3 LOTs starting with 1L Nivo+Ipi cohort and 39.7 months [95% CI: 30.4, NR] for the ≥2 LOTs starting with 1L IO+Axi cohort [**Figure 2**]
 - Numerical differences between the two cohorts were observed across IMDC risk groups [**Figure 3-5**]
 - Landmark analyses at 12, 18, and 24 months showed numerically higher median OS for the ≥2 LOTs starting with 1L IO+Axi cohort than the ≥3 LOTs starting with 1L Nivo+Ipi cohort at each landmark time point
 - A sensitivity analysis of patients in the ≥2 LOTs starting with 1L IO+Axi cohort who went on to receive any 3L (N=41) found consistent median OS at 39.7 months [95% CI: 25.3, NR]
 - After adjusting for potential confounders, there was no statistically significant difference in the hazard of death between cohorts as a whole; however statistical differences were observed between IMDC risk groups [**Table 3**]

LIMITATIONS

- Unmeasured confounding could account for associations observed in this study. We attempted to reduce bias by adjusting for potential confounders including age, sex, number of sites of metastases, lymph node metastases, and IMDC risk group
- There may be selection bias, as this study only included patients who received ≥3 LOTs starting with 1L Nivo+Ipi or ≥2 LOTs starting with 1L IO+Axi. These analyses may have excluded patients who had prolonged responses to 1L or died prior to receiving the required LOTs. However, consistent results from the sensitivity analyses help address the potential bias and support our findings of survival between the two cohorts being similar
- Treatment selection patterns may vary based on reimbursement factors unique to the region of clinical practice, which may not be representative
- Missing data exist in the IMDC database, which may bias study results if there is an underlying reason for missingness (i.e., missingness is not completely random)
- OS analysis was designed to be hypothesis generating and was not powered to detect statistically significant differences in survival between the two cohorts

OBJECTIVE

To assess the impact of treatment with ≥3 LOTs starting with 1L Nivo+Ipi and ≥2 LOTs starting with 1L IO+Axi on overall survival (OS)

CONCLUSIONS

- Our results demonstrate that the median OS was 33.0 months [95% CI: 27.6, 40.2] for patients who received ≥3 LOTs starting with 1L Nivo+Ipi and 39.7 months [95% CI: 30.4, NR] for patients who received ≥2 LOTs starting with 1L IO+Axi in a real-world database analysis
- Although future studies with larger sample sizes are warranted to compare clinical outcomes between the two cohorts, this study suggests that selection of effective treatments in 1L resulting in fewer LOTs may have similar clinical outcomes as multiple LOTs

Please scan this quick response (QR) code with your smartphone app to view an electronic version of this poster. If you do not have a smartphone, access the poster via the Internet at: https://meetings.asco.org/abstracts-presentations/216665

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission from ASCO® or the authors of this poster.

Table 1. Baseline Demographics and Clinical Characteristics among Patients with mRCC who Received 1L IO Combination Treatment and Subsequent Treatments[1]

	≥3 LOTs starting with 1L Nivo+Ipi N = 128	≥2 LOTs starting with 1L IO+Axi N = 104	p-value
Demographic Characteristics			
Age at index, years	*122*	*103*	
Mean ± SD	59.9 ± 10	61.6 ± 10.5	0.27
Median (IQR)	61.0 (55.1, 65.8)	62.0 (54.1, 68.8)	
Race, n (%)	*95*	*84*	
White	73 (76.8)	63 (75.0)	0.77
Sex, n (%)	*128*	*104*	
Male	98 (76.6)	70 (67.3)	0.12
Female	30 (23.4)	34 (32.7)	
Region, n (%)	*128*	*104*	
US	47 (36.7)	39 (37.5)	0.90
Non-US	81 (63.3)	65 (62.5)	
Tumor Characteristics			
Pathology, n (%)	*110*	*97*	
Clear cell	95 (86.4)	84 (86.6)	0.96
Non-clear cell	15 (13.6)	13 (13.4)	
Number of sites of metastases, n (%)	*119*	*101*	
1	36 (30.3)	15 (14.9)	< 0.01 *
> 1	83 (69.7)	86 (85.1)	
Location of metastases, n (%)	*117*	*100*	
Lung	79 (67.5)	69 (69.0)	0.82
Lymph nodes	48 (41.0)	67 (67.0)	< 0.01 *
Bone	37 (31.6)	36 (36.0)	0.50
Liver	25 (21.4)	16 (16.0)	0.31
Adrenal gland	15 (12.8)	15 (15.0)	0.64
Pancreas	9 (7.7)	12 (12.0)	0.28
Brain	4 (3.4)	4 (4.0)	1.00
Other[2]	26 (22.2)	34 (34.0)	0.05
Clinical Characteristics			
IMDC risk group, n (%)	*109*	*94*	
Favorable	15 (13.8)	26 (27.7)	0.01 *
Intermediate	66 (60.6)	45 (47.9)	0.07
Poor	28 (25.7)	23 (24.5)	0.84
Prior nephrectomy, n (%)	*128*	*104*	
Yes	89 (69.5)	83 (79.8)	0.08
No	39 (30.5)	21 (20.2)	

Abbreviations: 1L: first line; IMDC: International Metastatic Renal Cell Carcinoma Database Consortium; IO: immune-oncologic; IO+Axi; immuno-oncology+Axitinib; IQR: interquartile range; LOT: line of therapy; mRCC: metastatic renal cell carcinoma; Nivo+Ipi, Nivolumab+Ipilimumab; standard deviation; US: United States

Notes:

*Indicates p-value <0.05.

[1] The baseline period was defined as the time from mRCC diagnosis to 1L IO combination treatment initiation.

[2] Other metastases included metastases on bowel, kidney, pelvis, peritoneum, pleura, soft tissue, spleen, and/or thyroid.

Figure 1. Sankey Diagrams: 2L and 3L Treatment Sequences among Patients with mRCC who received 1L IO Combination Treatment and Subsequent Treatments[1]

≥3 LOTs starting with 1L Nivo+Ipi

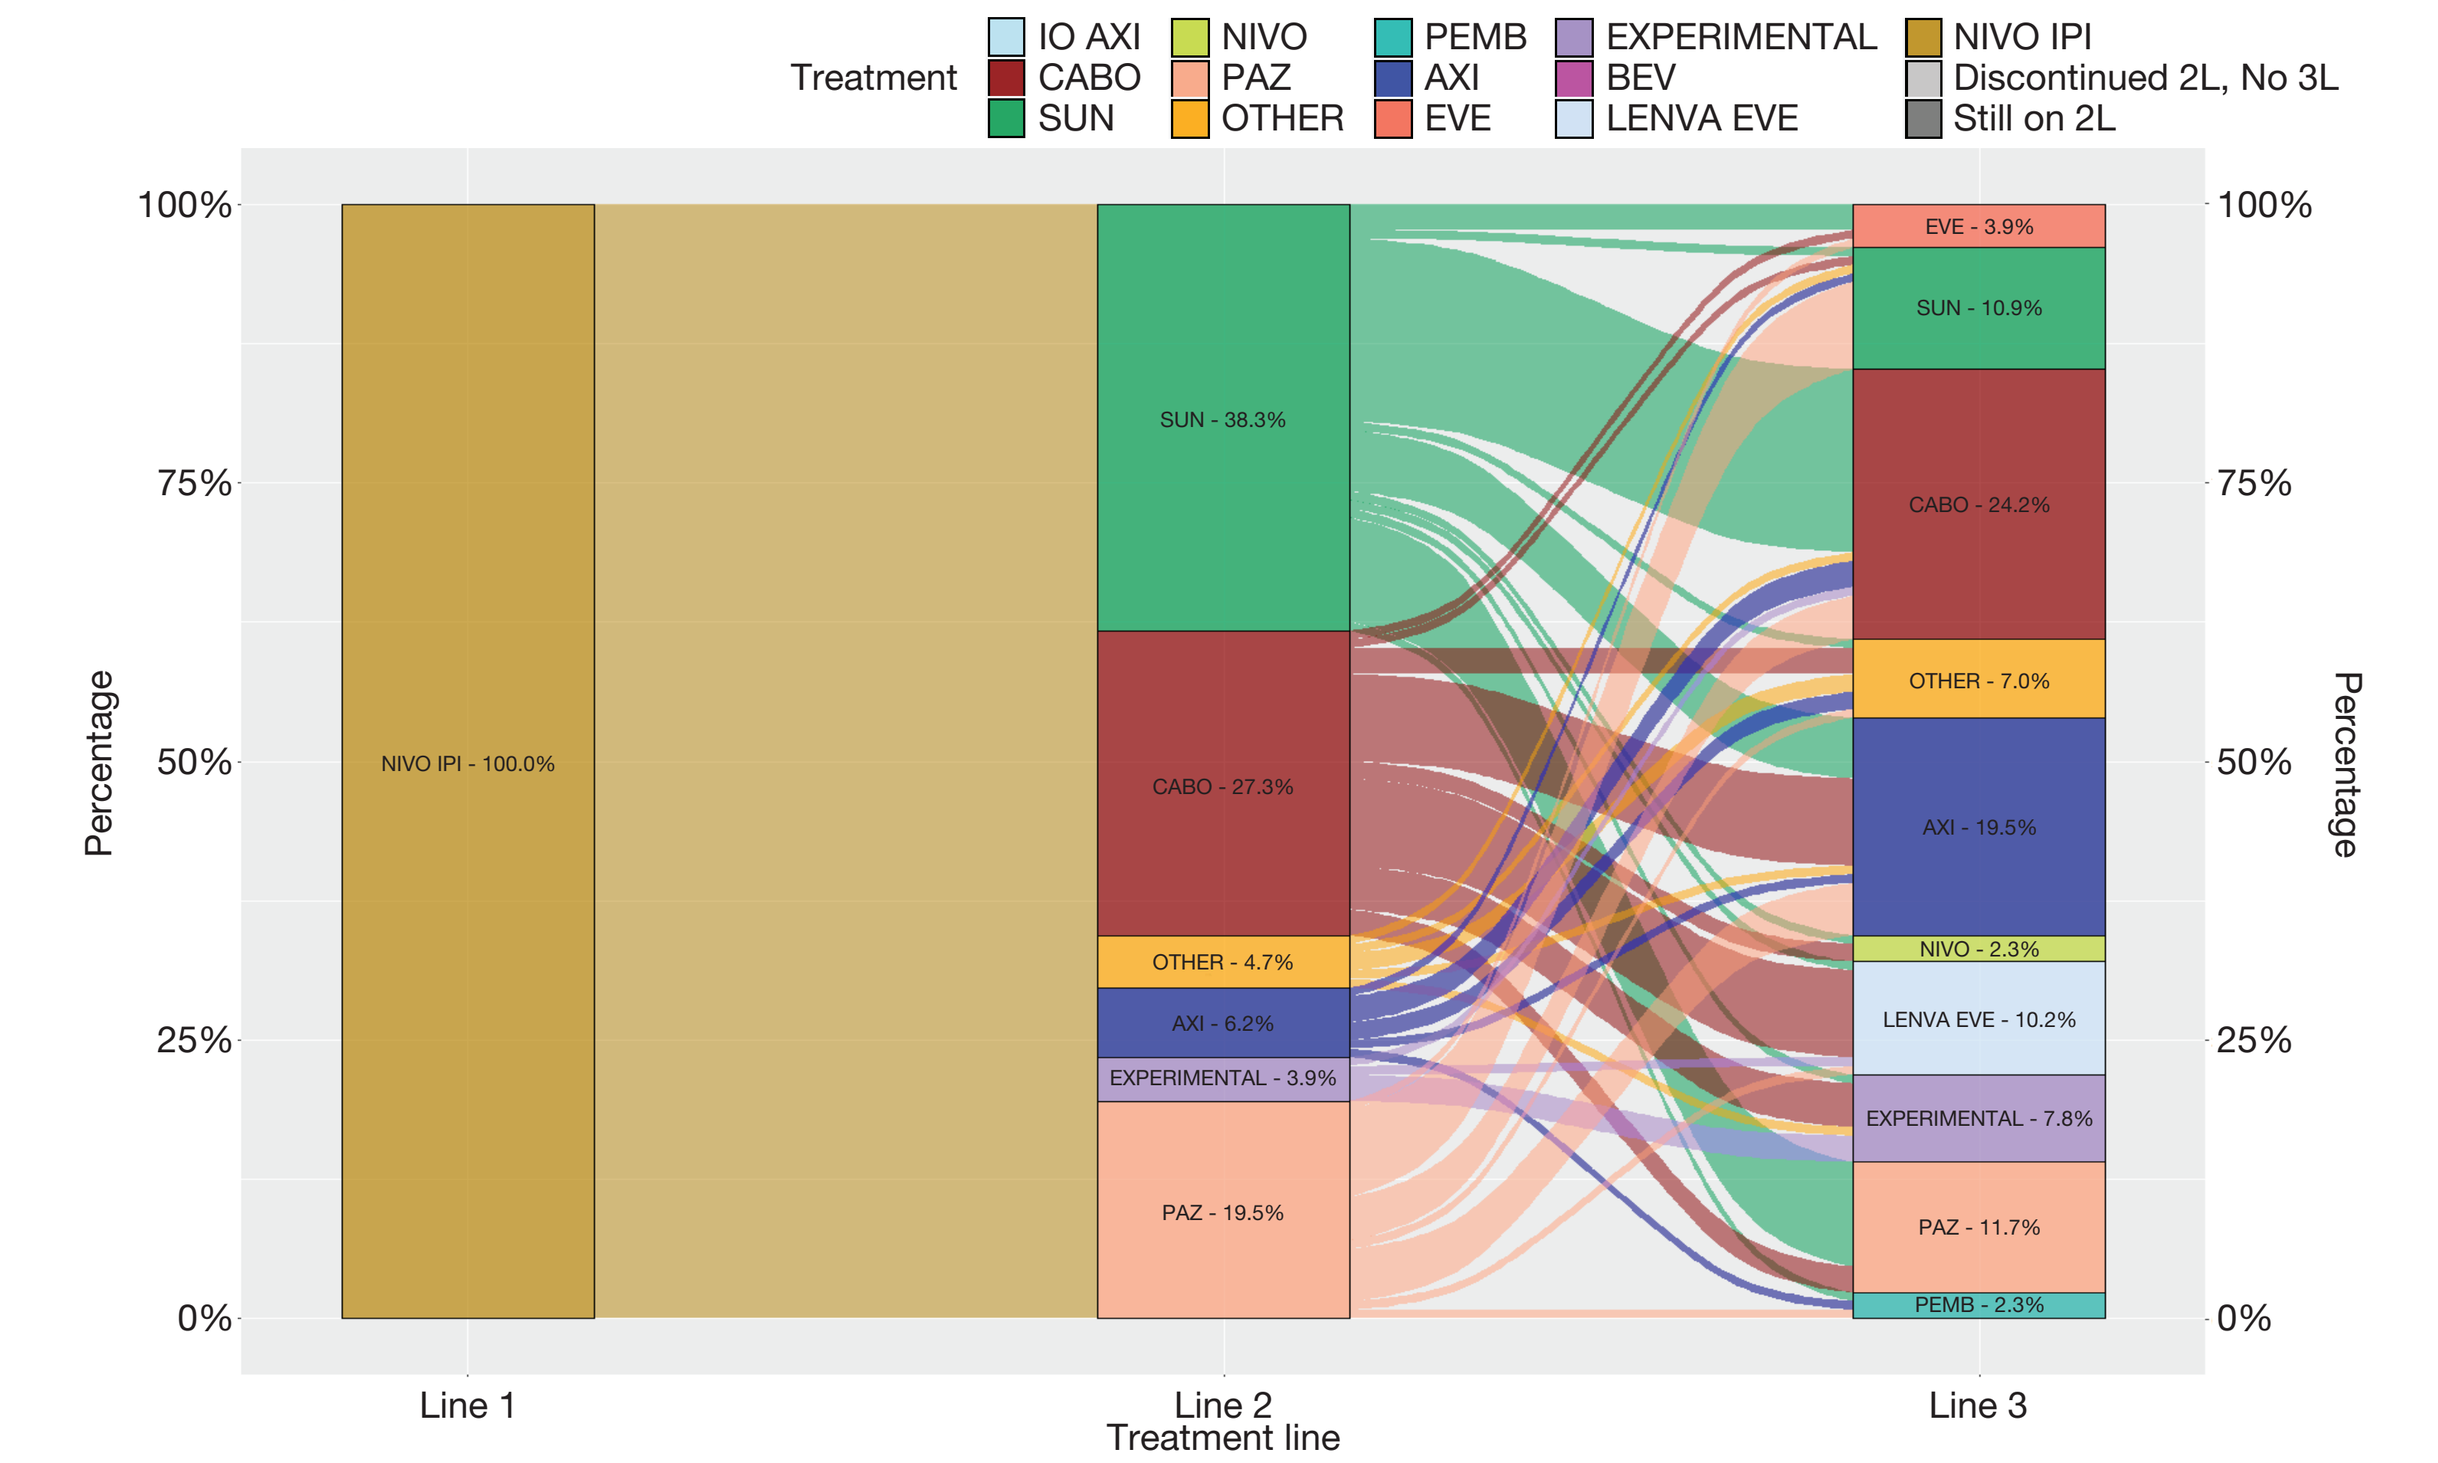
Treatment
IO AXI
CABO
SUN
NIVO
PAZ
OTHER
PEMB
AXI
EVE
EXPERIMENTAL
BEV
LENVA EVE
NIVO IPI
Discontinued 2L, No 3L
Still on 2L
Percentage
100%
75%
50%
25%
0%
NIVO IPI - 100.0%
SUN - 38.3%
CABO - 27.3%
OTHER - 4.7%
AXI - 6.2%
EXPERIMENTAL - 3.9%
PAZ - 19.5%
EVE - 3.9%
SUN - 10.9%
CABO - 24.2%
OTHER - 7.0%
AXI - 19.5%
NIVO - 2.3%
LENVA EVE - 10.2%
EXPERIMENTAL - 7.8%
PAZ - 11.7%
PEMB - 2.3%
Line 1
Line 2
Line 3
Treatment line

≥2 LOTs starting with 1L IO+Axi

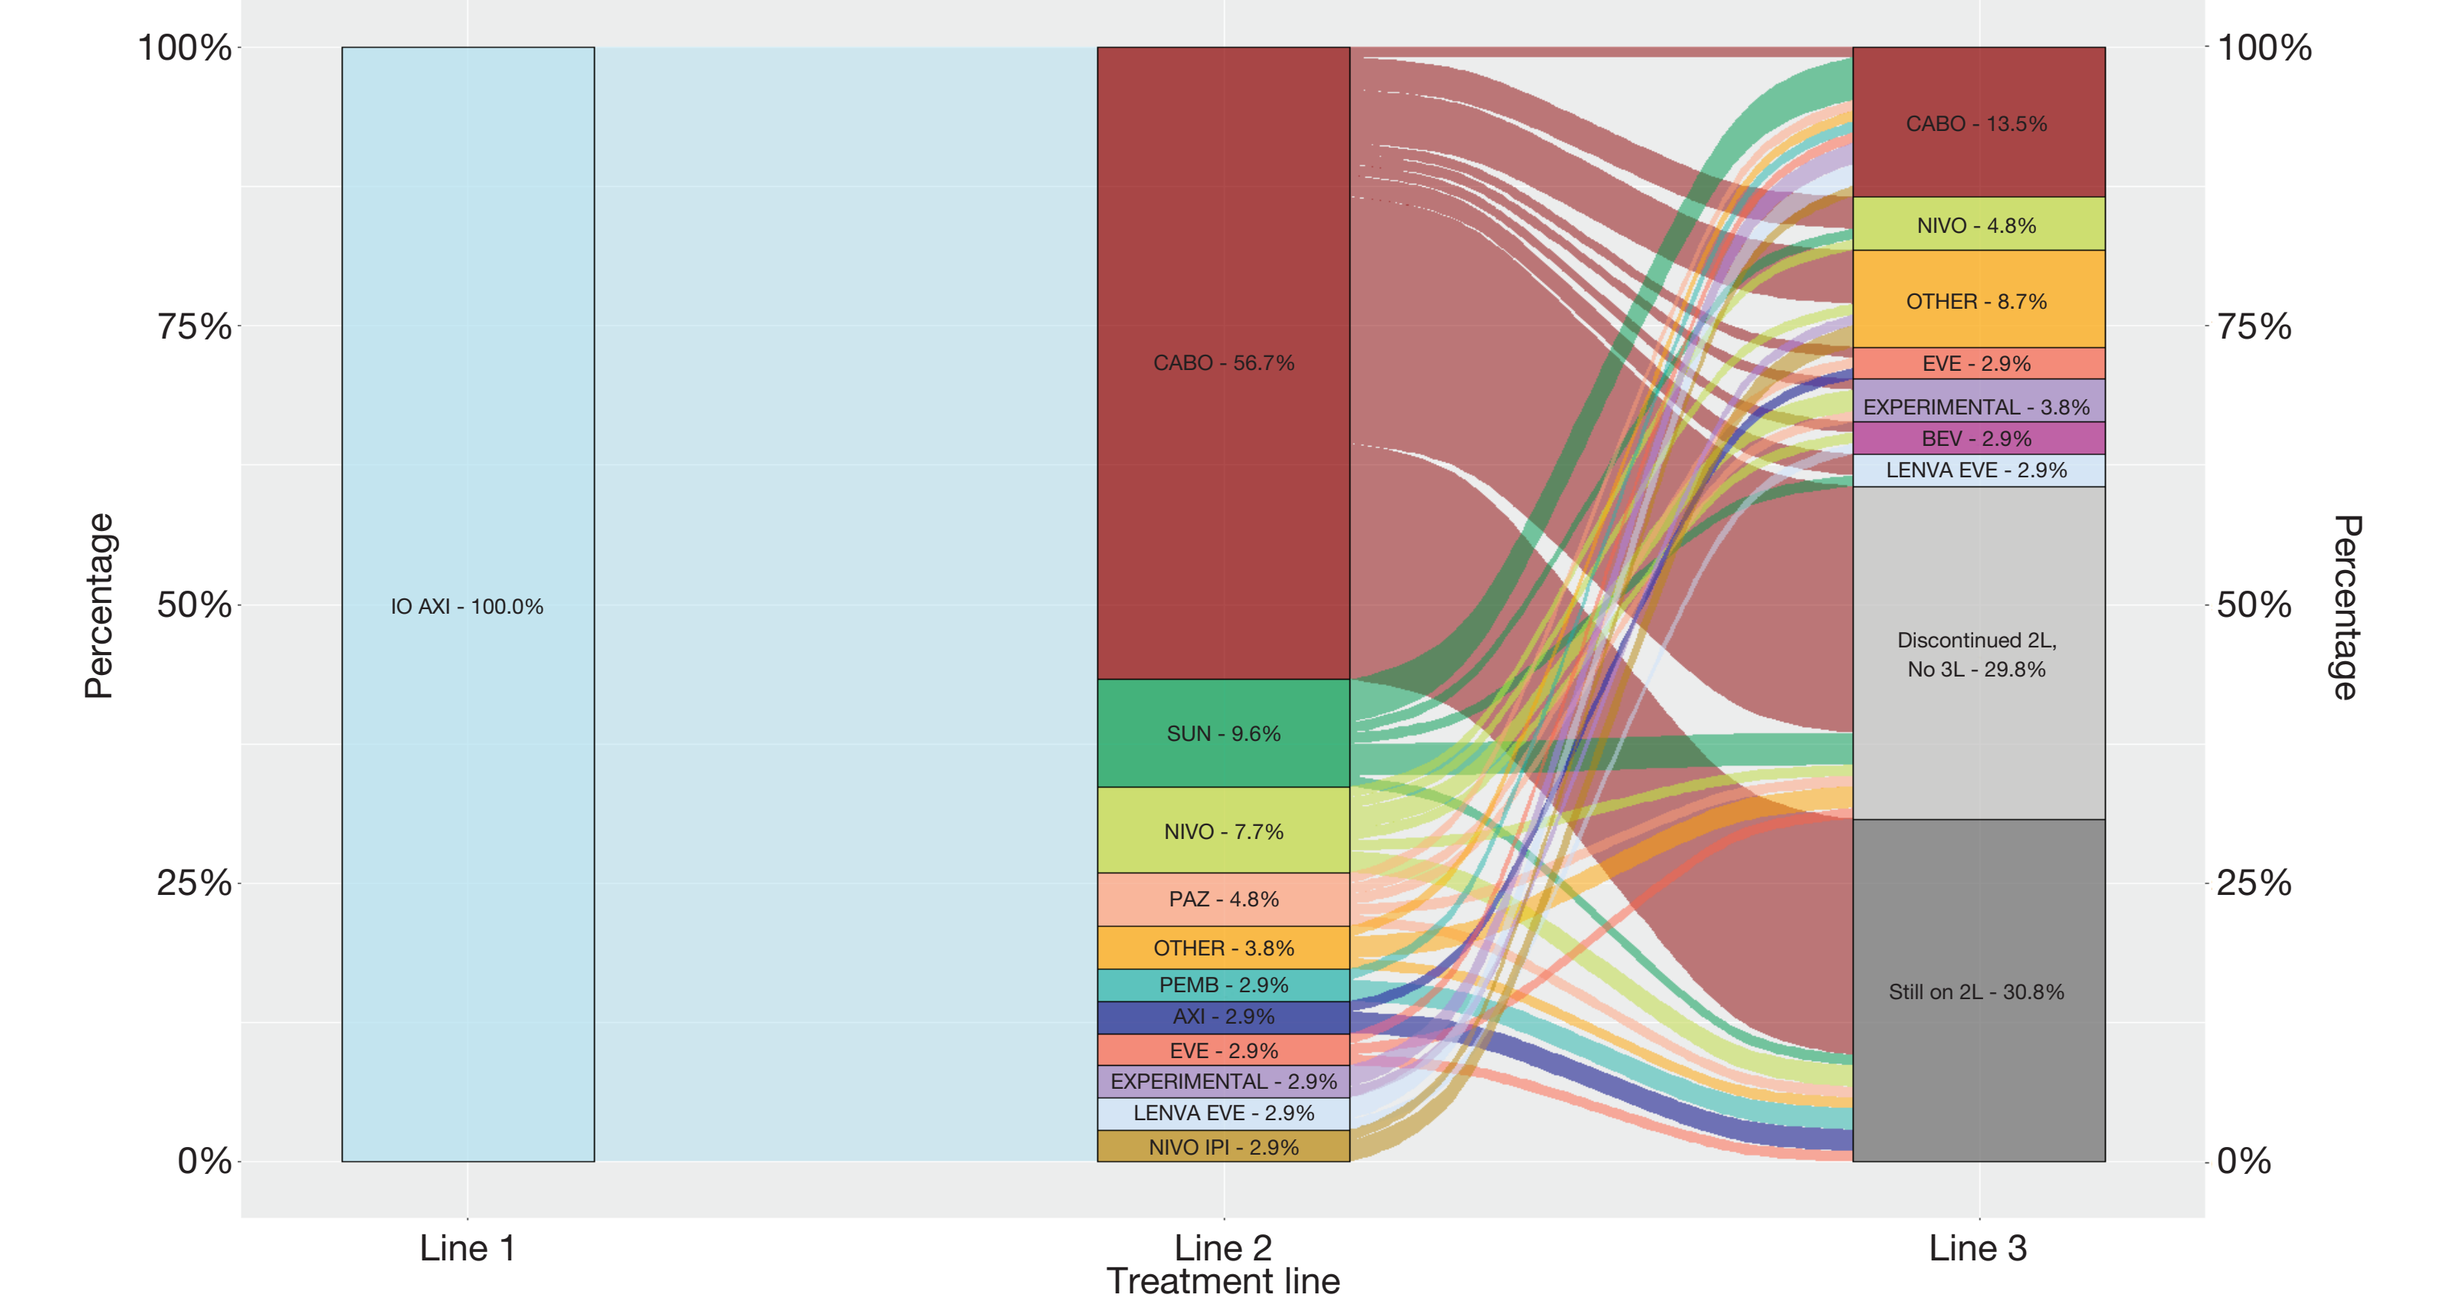
Percentage
100%
75%
50%
25%
0%
IO AXI - 100.0%
CABO - 56.7%
SUN - 9.6%
NIVO - 7.7%
PAZ - 4.8%
OTHER - 3.8%
PEMB - 2.9%
AXI - 2.9%
EVE - 2.9%
EXPERIMENTAL - 2.9%
LENVA EVE - 2.9%
NIVO IPI - 2.9%
CABO - 13.5%
NIVO - 4.8%
OTHER - 8.7%
EVE - 2.9%
EXPERIMENTAL - 3.8%
BEV - 2.9%
LENVA EVE - 2.9%
Discontinued 2L, No 3L - 29.8%
Still on 2L - 30.8%
Line 1
Line 2
Line 3
Treatment line

Abbreviations: 1L, first-line; 2L, second-line; 3L, third-line; AXI, axitinib; BEV, bevacizumab; CABO, cabozantinib; EVE, everolimus; IO, immuno-oncology; IO AXI, immuno-oncology + axitinib; LENVA, lenvatinib; LENVA EVE, lenvatinib + everolimus; LOT: line of therapy; mRCC, metastatic renal cell carcinoma; NIVO, nivolumab; NIVO IPI, Nivolumab + Ipilimumab; PAZ, Pazopanib; PEMB, Pembrolizumab; SUN, Sunitinib

Note:

[1] Other represents a combined category of treatments that have N < 3

Table 2. TTD and TTNT by Treatment Line Among Patients with mRCC Who Received 1L IO Combination Treatment and Subsequent Treatments

	≥3 LOTs starting with 1L Nivo+Ipi		≥2 LOTs starting with 1L IO+Axi	
	N	Median Time to Event [95% CI], months	N	Median Time to Event [95% CI], months
Duration of Follow-up				
Overall	128	21.6 [19.9, 24.9]	104	22.0 [18.9, 25.2]
TTD				
1L	128	3.1 [2.7, 3.9]	104	8.2 [6.5, 9.8]
2L	128	4.1 [3.1, 5.1]	104	6.1 [4.8, 8.4]
3L	128	4.7 [3.5, 5.9]	41	6.2 [3.1, 7.5]
TTNT				
1L	128	5.8 [3.9, 6.9]	104	8.7 [7.0, 11.5]
2L	128	4.6 [3.9, 5.7]	104	12.6 [10.1, 25.1]

Abbreviations: 1L, first-line; 2L, second-line; 3L, third-line; Axi, axitinib; CI, confidence interval; IO, immuno-oncology; IO+Axi, immuno-oncology+Axitinib; LOT: line of therapy; mRCC, metastatic renal cell carcinoma; NA, not applicable; Nivo+Ipi, Nivolumab + Ipilimumab; TTD, time to treatment discontinuation; TTNT, time to next treatment; NR, not reached

Table 3. Overall Survival Among Patients with mRCC who Received 1L IO Combination Treatment and Subsequent Treatments

	Overall Survival: HR [95% CI][1]	
	Unadjusted	Adjusted
≥2 LOTs starting with 1L IO+Axi cohort (ref: ≥3 LOTs starting with 1L Nivo+Ipi cohort)	0.68 [0.45, 1.02]	0.82 [0.50, 1.32]
Age at index date ≥ 60 years (ref: age at index date < 60 years)	-	1.04 [0.66, 1.65]
Sex (ref: male)	-	0.72 [0.41, 1.25]
1L IMDC risk group (ref: 1L Poor IMDC risk group)		
1L Favorable IMDC risk group	-	0.31 [0.16, 0.62] *
1L Intermediate IMDC risk group	-	0.36 [0.20, 0.62] *
≥ 1 site of metastases (ref: 1 site of metastasis)	-	1.29 [0.71, 2.34]
Lymph node metastases (ref: no lymph node metastases)	-	1.09 [0.67, 1.78]

Abbreviations: 1L, first-line; 2L, second-line; 3L, third-line; Axi, axitinib; HR, hazard ratio; CI, confidence interval; IO, immuno-oncology; IO+Axi, immuno-oncology+Axitinib; LOT: line of therapy; mRCC, metastatic renal cell carcinoma; Nivo+Ipi, Nivolumab + Ipilimumab; OS, overall survival

*Indicates p-value <0.05.

Notes:

[1] HR<1 indicates patients with the characteristic of interest had lower hazard of death (i.e., favorable OS) compared to patients in the reference group. In this study, the ≥2 LOTs starting with 1L IO+Axi cohort had no statistically significant difference in OS compared to the ≥3 LOTs starting with 1L Nivo+Ipi cohort.

Figure 2. Kaplan-Meier Analysis of Overall Survival for Patients with mRCC who Received 1L IO Combination Treatment and Subsequent Treatments[1]

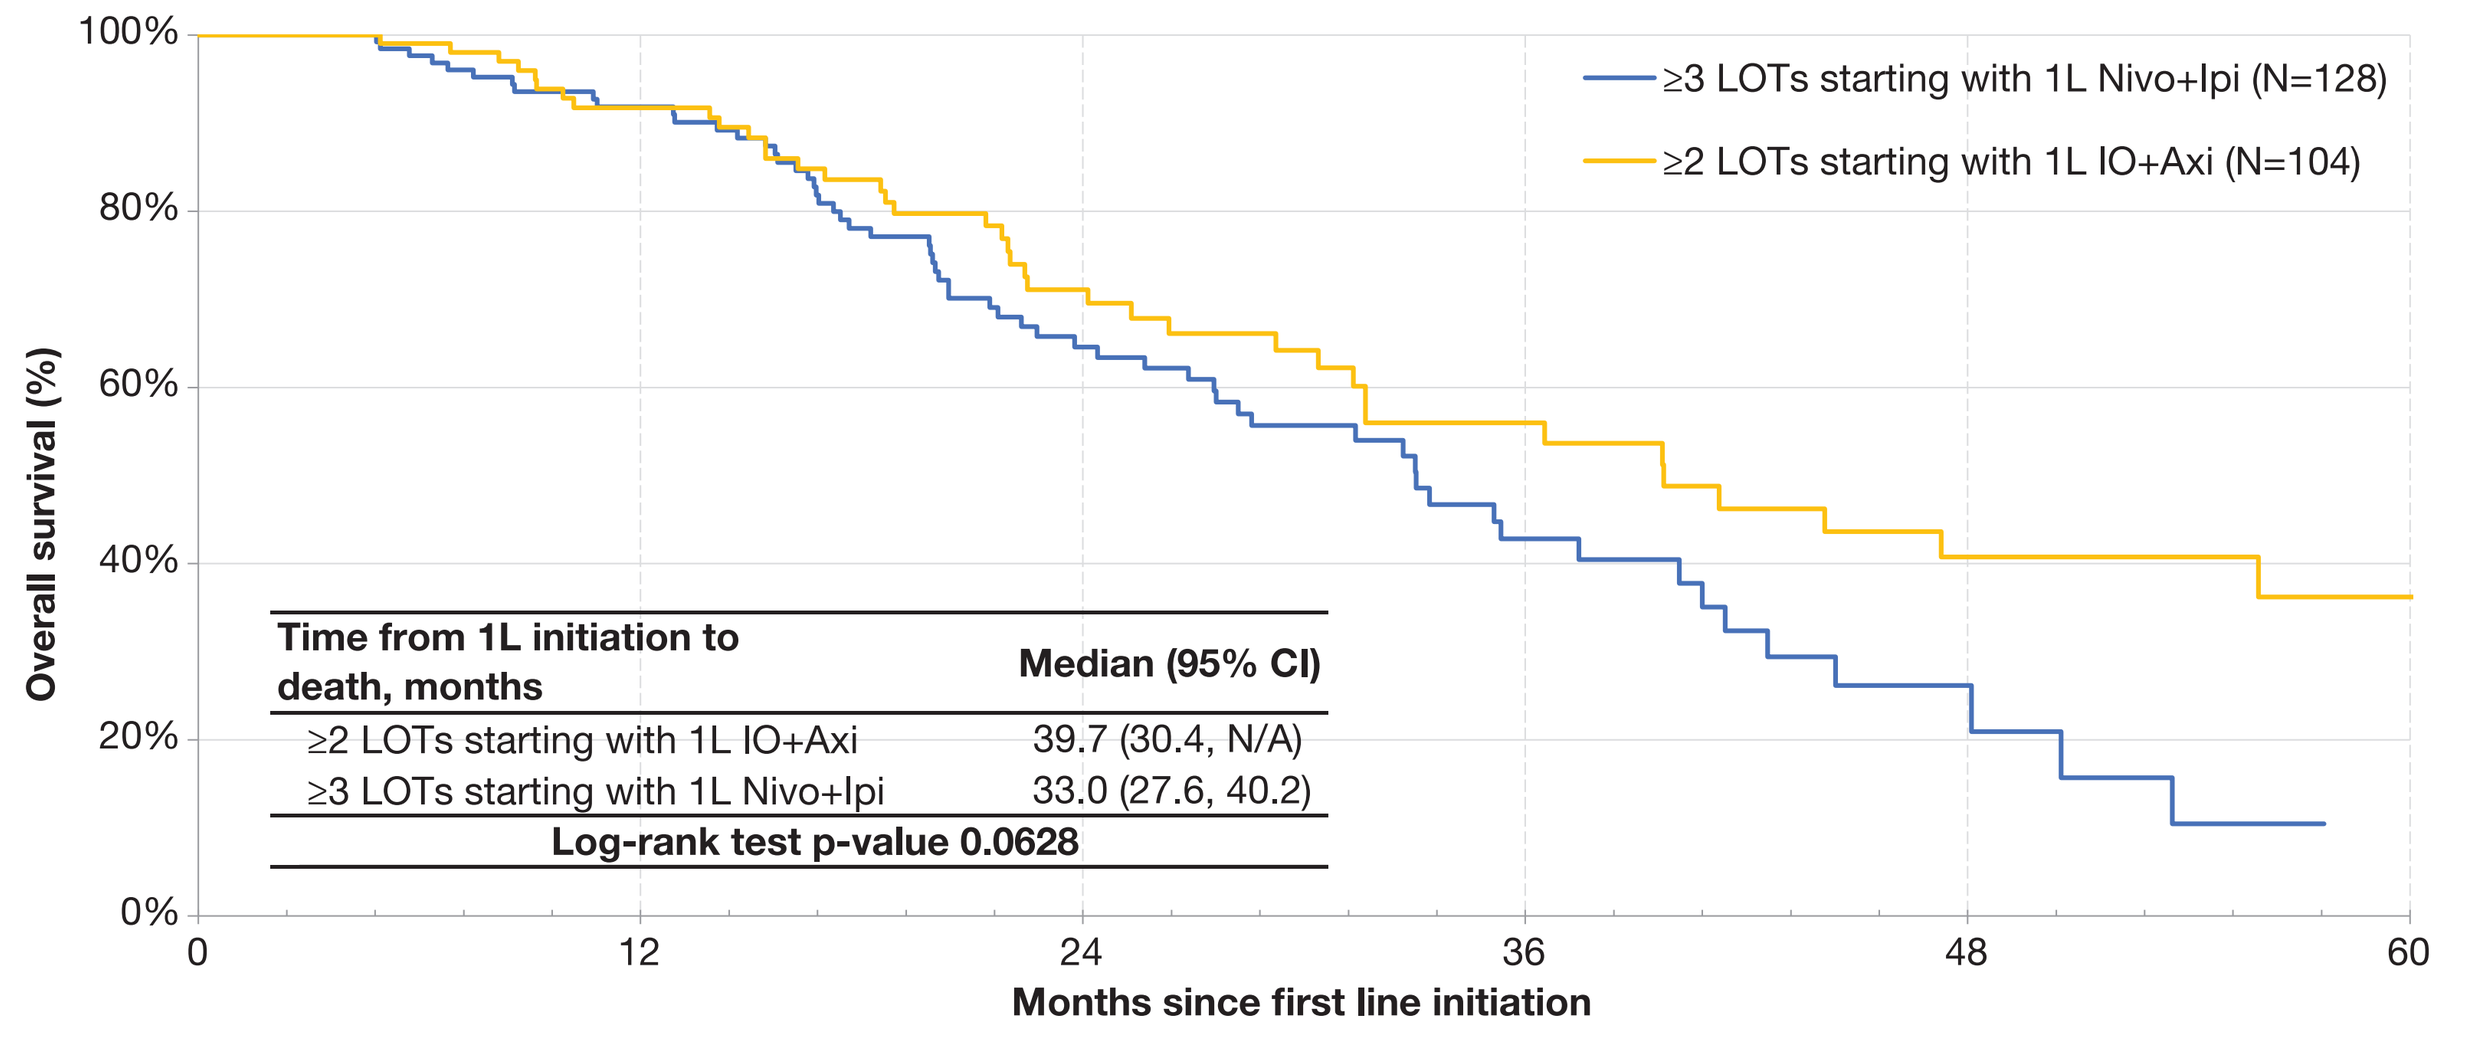
≥3 LOTs starting with 1L Nivo+Ipi (N=128)
≥2 LOTs starting with 1L IO+Axi (N=104)
Overall survival (%)
100%
80%
60%
40%
20%
0%
Time from 1L initiation to death, months
Median (95% CI)
≥2 LOTs starting with 1L IO+Axi 39.7 (30.4, N/A)
≥3 LOTs starting with 1L Nivo+Ipi 33.0 (27.6, 40.2)
Log-rank test p-value 0.0628
0
12
24
36
48
60
Months since first line initiation

Abbreviations: 1L, first-line; 2L, second-line; 3L, third-line; Axi, axitinib; CI, confidence interval; IO, immuno-oncology; IO+Axi, immuno-oncology+Axitinib; Nivo+Ipi, Nivolumab + Ipilimumab; LOT: line of therapy; mRCC, metastatic renal cell carcinoma

Notes:

[1] Over the entire study period, 39 (37.5%) of patients in ≥2 LOTs starting with 1L IO+Axi cohort, and 61 (47.7%) patients in the ≥3 LOTs starting with 1L Nivo+Ipi cohort had the event.

Figure 3. Kaplan-Meier Analysis of Overall Survival for Patients with mRCC who Received 1L IO Combination Treatment and in Favorable IMDC Risk Group, Stratified by Treatment Sequence[1]

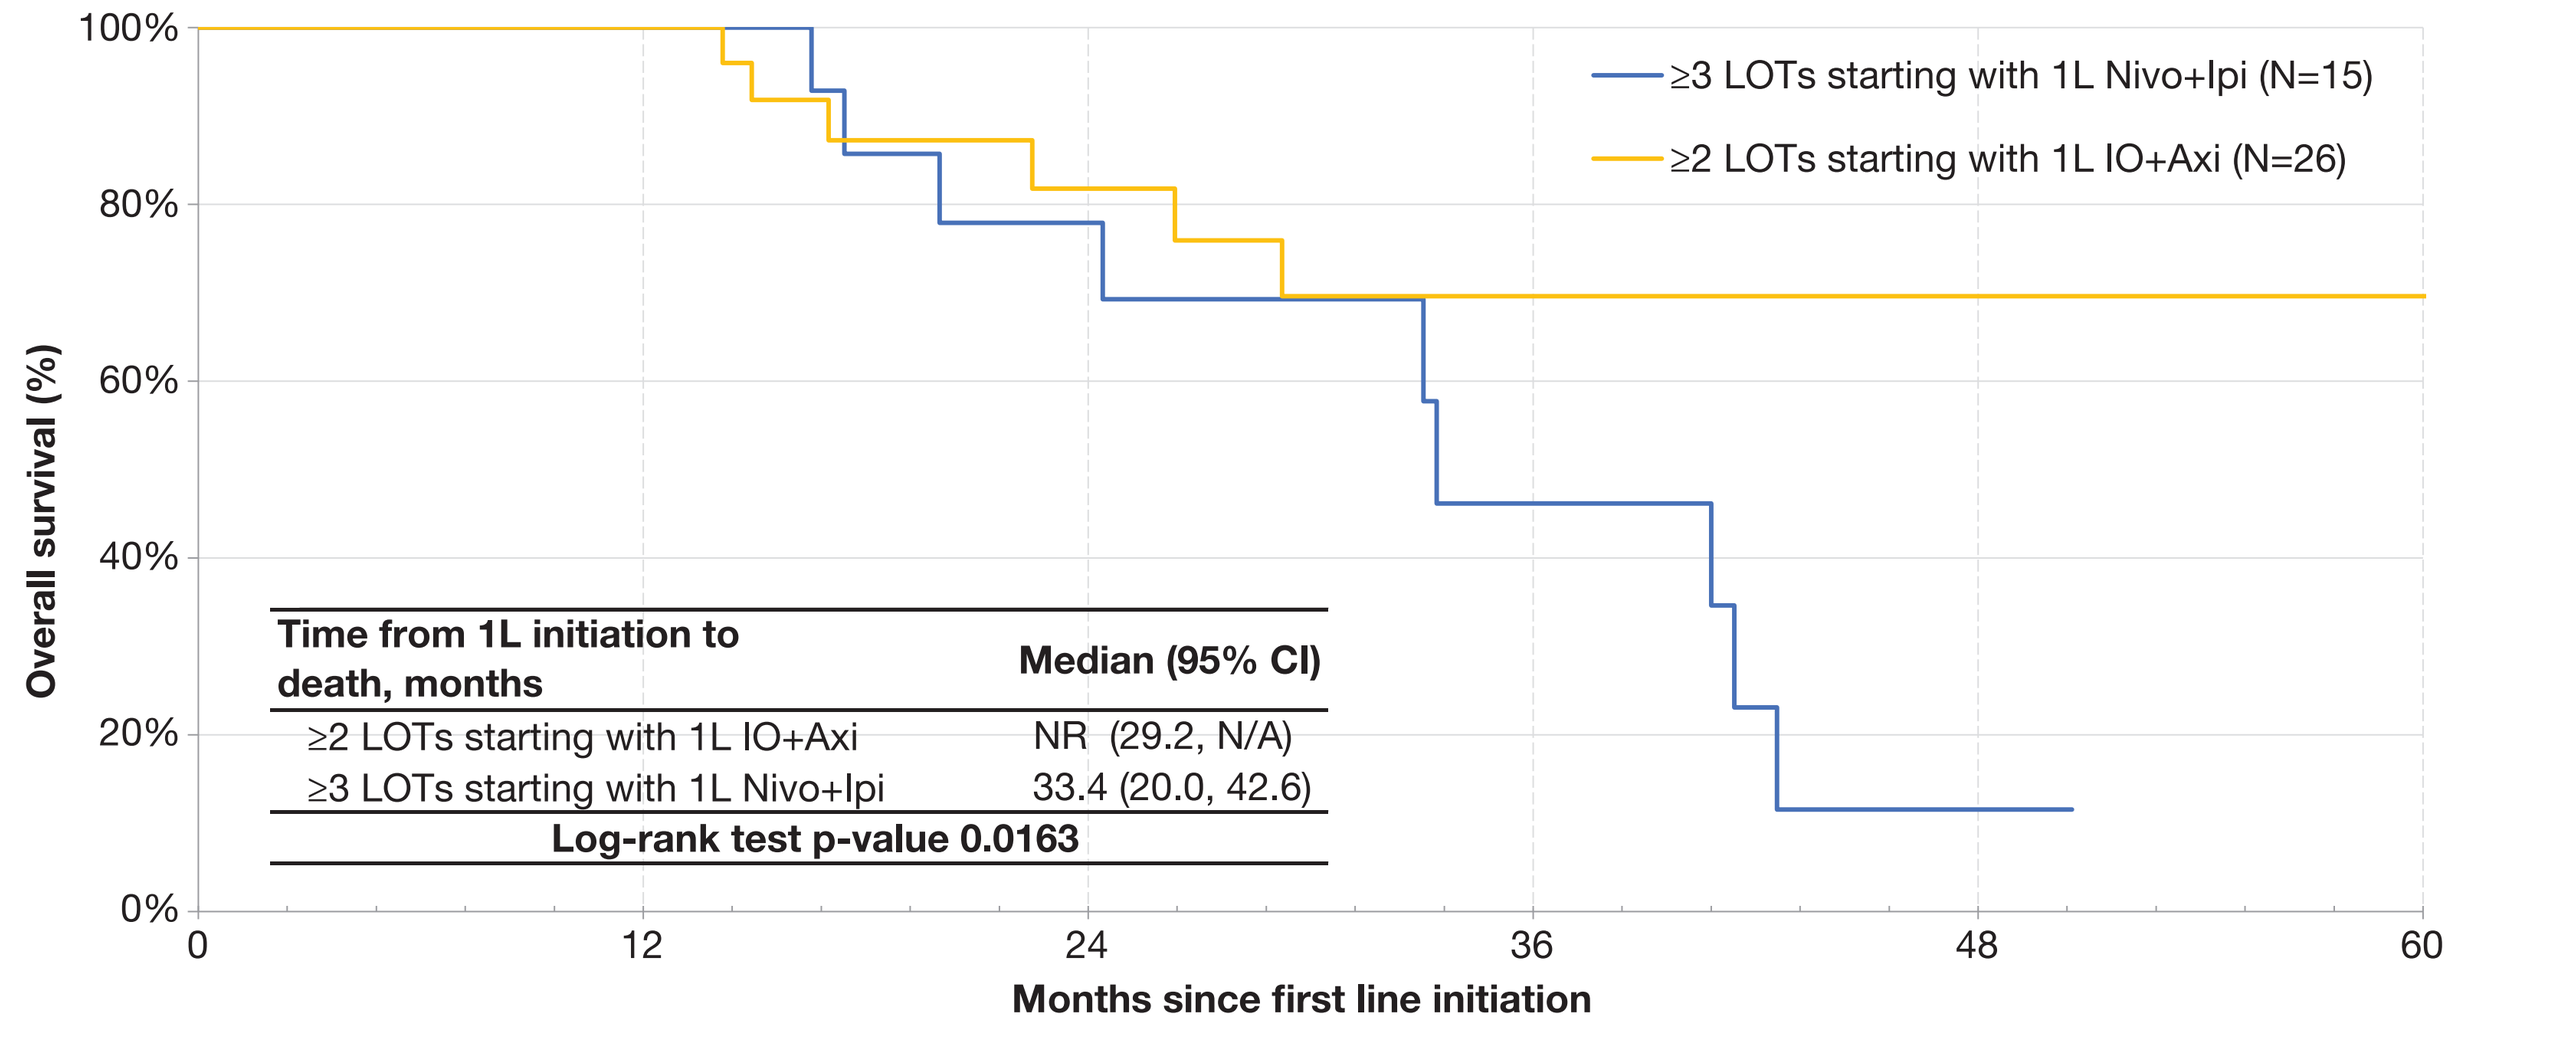
≥3 LOTs starting with 1L Nivo+Ipi (N=15)
≥2 LOTs starting with 1L IO+Axi (N=26)
Overall survival (%)
100%
80%
60%
40%
20%
0%
Time from 1L initiation to death, months
Median (95% CI)
≥2 LOTs starting with 1L IO+Axi NR (29.2, N/A)
≥3 LOTs starting with 1L Nivo+Ipi 33.4 (20.0, 42.6)
Log-rank test p-value 0.0163
0
12
24
36
48
60
Months since first line initiation

Abbreviations: 1L, first-line; 2L, second-line; 3L, third-line; Axi, axitinib; CI, confidence interval; IO, immuno-oncology; IO+Axi, immuno-oncology+Axitinib; Nivo+Ipi, Nivolumab + Ipilimumab; LOT: line of therapy; mRCC, metastatic renal cell carcinoma

Notes:

[1] Over the entire study period, 6 (23.1%) of patients in ≥2 LOTs starting with 1L IO+Axi cohort, and 10 (66.7%) patients in the ≥3 LOTs starting with 1L Nivo+Ipi cohort had the event.

Figure 4. Kaplan-Meier Analysis of Overall Survival for Patients with mRCC who Received 1L IO Combination Treatment and in Intermediate IMDC Risk Group, Stratified by Treatment Sequence[1]

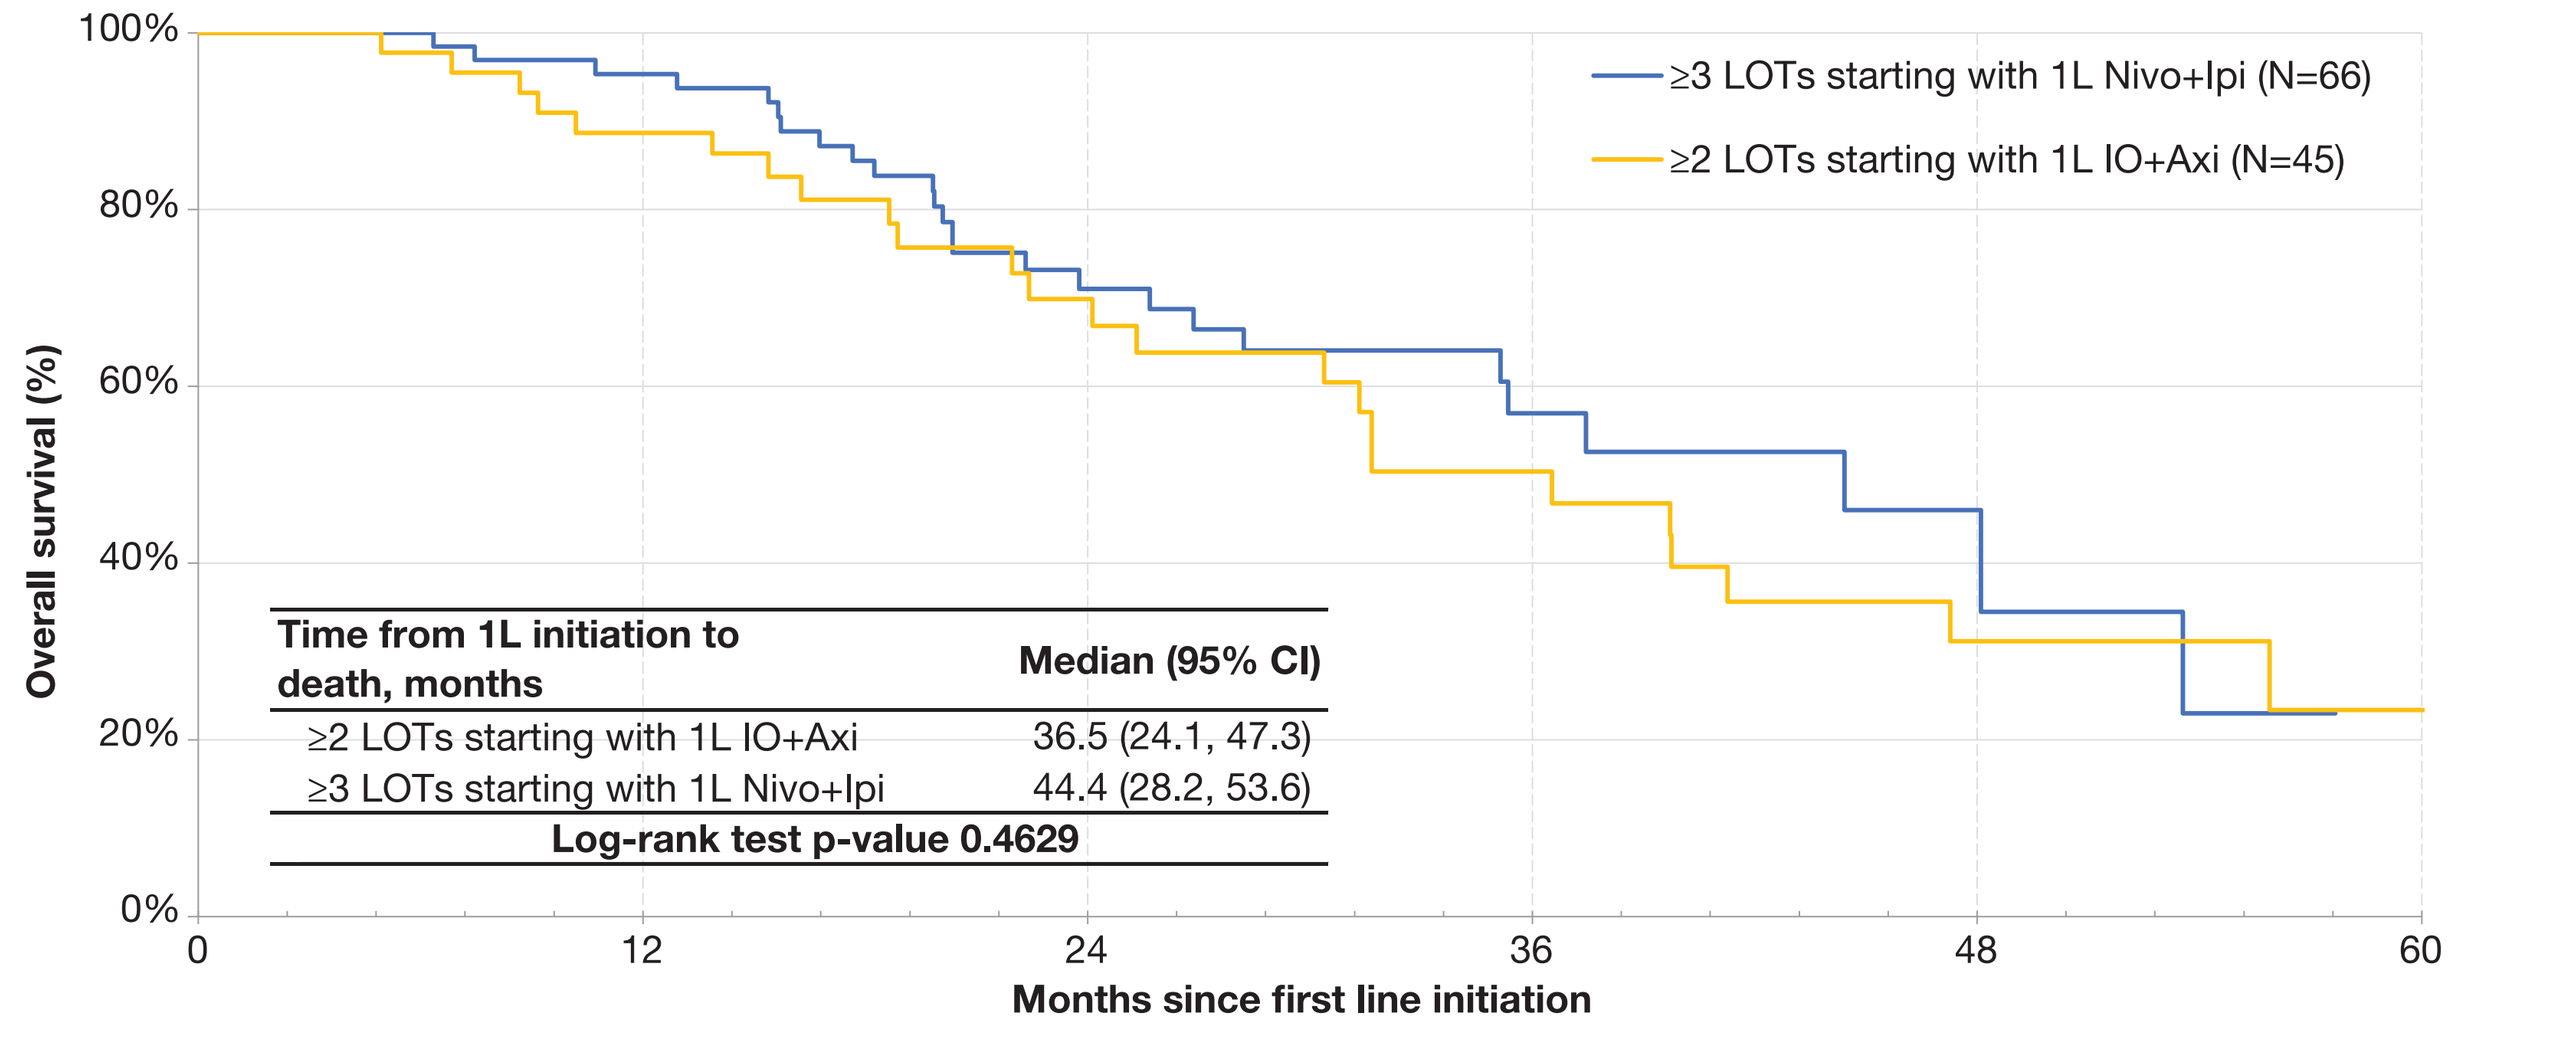
≥3 LOTs starting with 1L Nivo+Ipi (N=66)
≥2 LOTs starting with 1L IO+Axi (N=45)
Overall survival (%)
100%
80%
60%
40%
20%
0%
Time from 1L initiation to death, months
Median (95% CI)
≥2 LOTs starting with 1L IO+Axi 36.5 (24.1, 47.3)
≥3 LOTs starting with 1L Nivo+Ipi 44.4 (28.2, 53.6)
Log-rank test p-value 0.4629
0
12
24
36
48
60
Months since first line initiation

Abbreviations: 1L, first-line; 2L, second-line; 3L, third-line; Axi, axitinib; CI, confidence interval; IO, immuno-oncology; IO+Axi, immuno-oncology+Axitinib; Nivo+Ipi, Nivolumab + Ipilimumab; LOT: line of therapy; mRCC, metastatic renal cell carcinoma

Notes:

[1] Over the entire study period, 24 (53.3%) of patients in ≥2 LOTs starting with 1L IO+Axi cohort, and 26 (39.4%) patients in the ≥3 LOTs starting with 1L Nivo+Ipi cohort had the event.

Figure 5. Kaplan-Meier Analysis of Overall Survival for Patients with mRCC who Received 1L IO Combination Treatment and in Poor IMDC Risk Group, Stratified by Treatment Sequence[1]

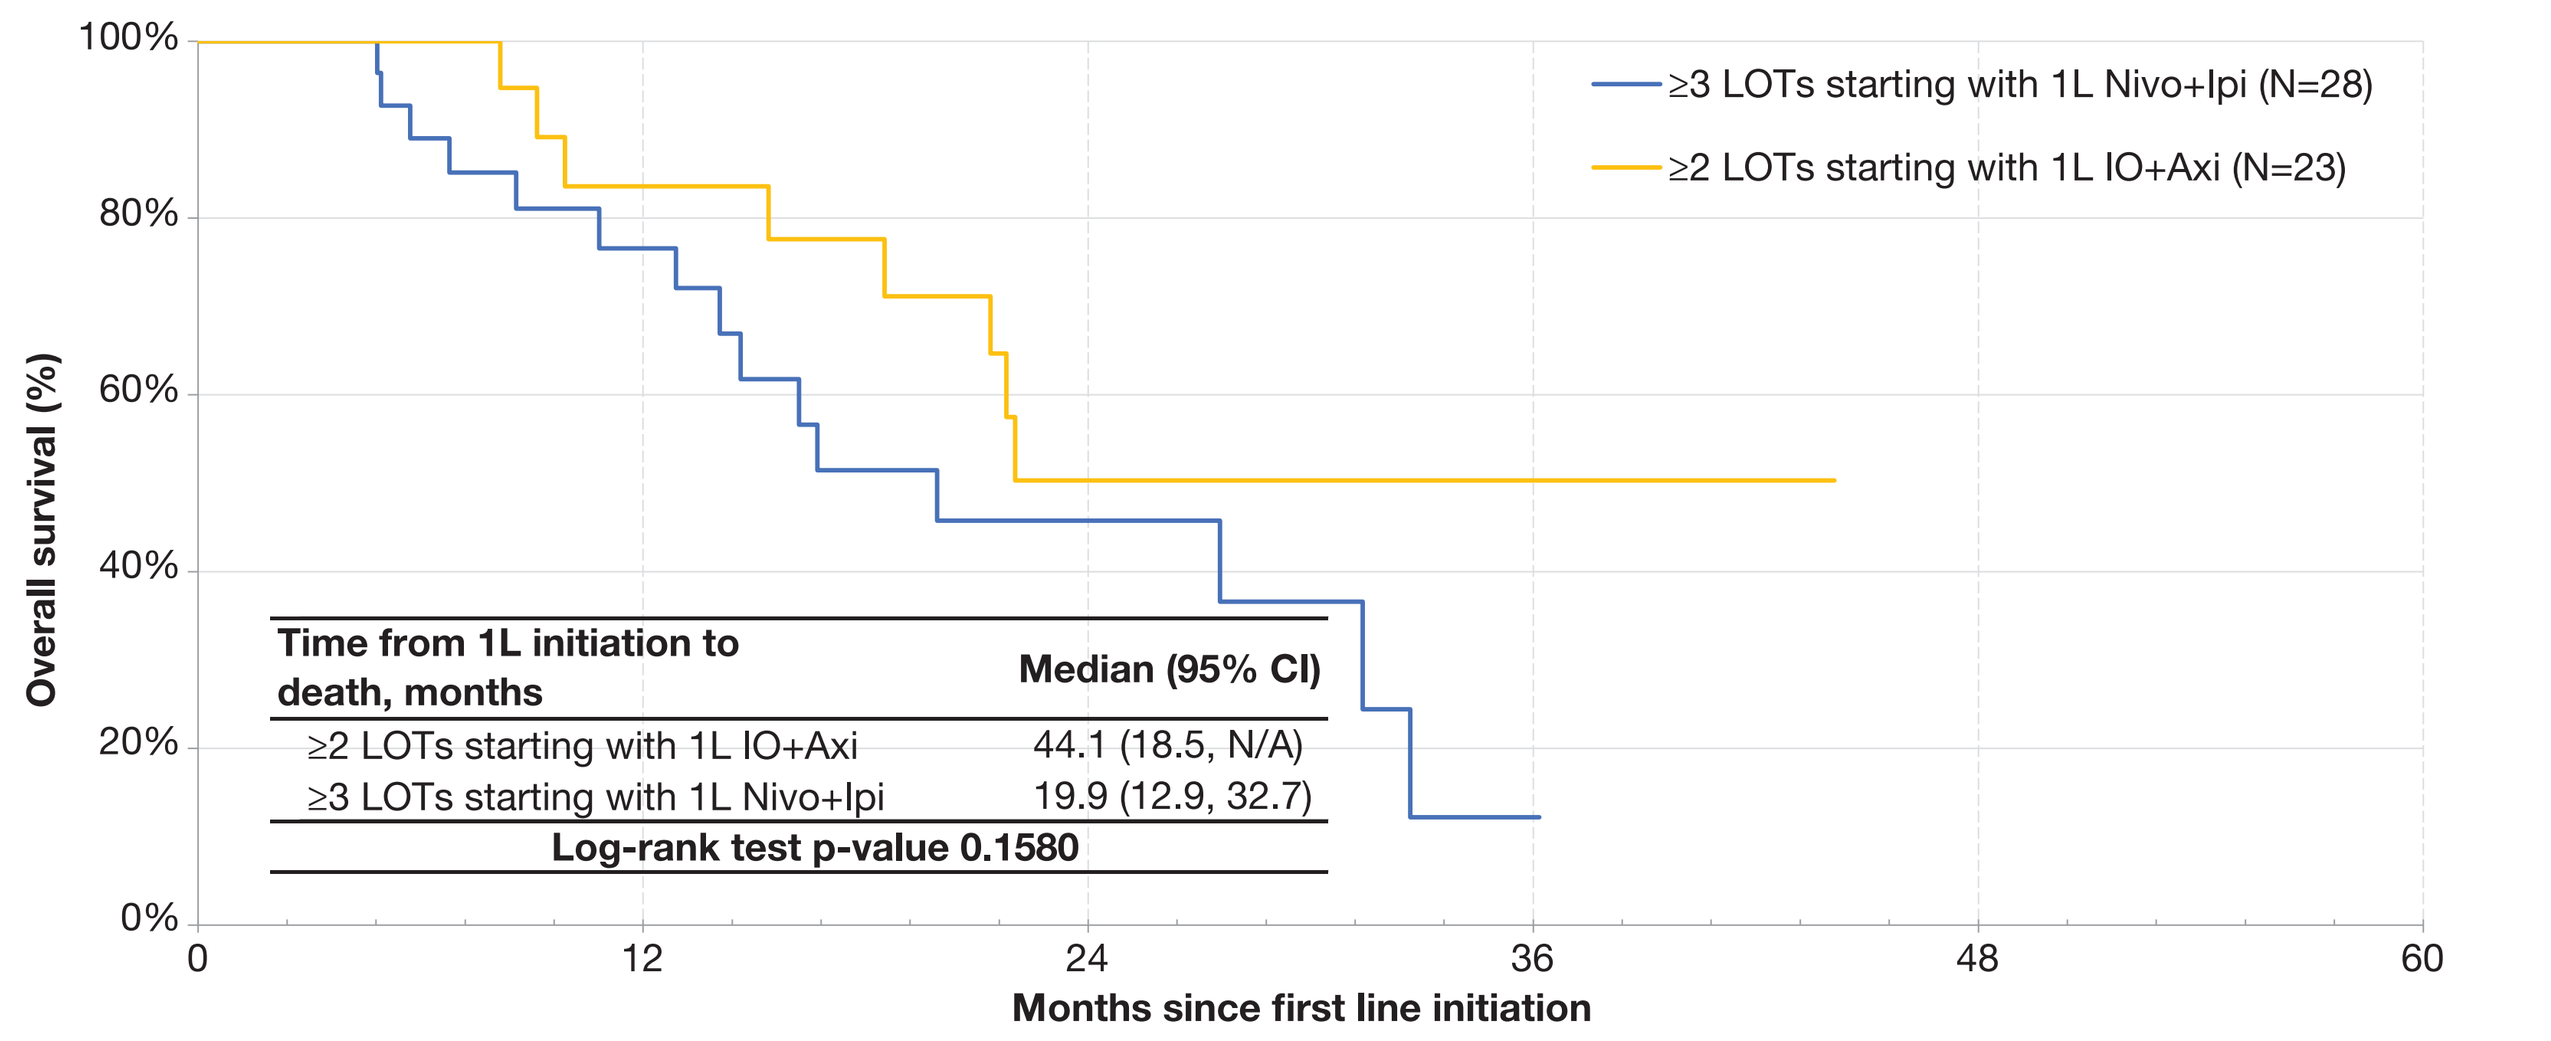
≥3 LOTs starting with 1L Nivo+Ipi (N=28)
≥2 LOTs starting with 1L IO+Axi (N=23)
Overall survival (%)
100%
80%
60%
40%
20%
0%
Time from 1L initiation to death, months
Median (95% CI)
≥2 LOTs starting with 1L IO+Axi 44.1 (18.5, N/A)
≥3 LOTs starting with 1L Nivo+Ipi 19.9 (12.9, 32.7)
Log-rank test p-value 0.1580
0
12
24
36
48
60
Months since first line initiation

Abbreviations: 1L, first-line; 2L, second-line; 3L, third-line; Axi, axitinib; CI, confidence interval; IO, immuno-oncology; IO+Axi, immuno-oncology+Axitinib; Nivo+Ipi, Nivolumab + Ipilimumab; LOT: line of therapy; mRCC, metastatic renal cell carcinoma

Notes:

[1] Over the entire study period, 9 (39.1%) of patients in ≥2 LOTs starting with 1L IO+Axi cohort, and 15 (53.6%) patients in the ≥3 LOTs starting with 1L Nivo+Ipi cohort had the event.

References

1. George, D. J., Lee, C. H., & Heng, D. (2021). New approaches to first-line treatment of advanced renal cell carcinoma. *Therapeutic advances in medical oncology*, 13, 17588359211034708. https://doi.org/10.1177/17588359211034708
2. Ernst, M. S., et al. (2022). Characterizing IMDC prognostic groups in contemporary first-line combination therapies for metastatic renal cell carcinoma (mRCC). *Journal of Clinical Oncology*. Vol. 40. No. 6.
3. Fitzgerald, K. N., et al. (2022). Progression-free Survival After Second Line of Therapy for Metastatic Clear Cell Renal Cell Carcinoma in Patients Treated with First-line Immunotherapy Combinations. *European Urology*. https://doi.org/10.1016/j.eururo.2022.10.017

Acknowledgements

This study was sponsored by Pfizer Inc.

Corresponding author: daniel.heng@albertahealthservices.ca

Presented at ASCO-GU, February 16-18, 2023, San Francisco, CA & Online